What is the relationship between price and prevalence in non-oncology rare disease? (Pro53)

Cecile Matthews, Bhavesh H Patel, Owen Male – CRA International (UK) Limited, 50/60 Station Road, Cambridge, United Kingdom

Introduction

High development costs and low patient numbers have often been presented as justifications for high treatment prices in rare diseases. Payers have traditionally accepted this narrative for two reasons. First, because the individual budget impact of each product remains low relative to high budget impact categories. And second, because of high unmet need and a regulatory environment that supports investment in orphan indications – often driven by broader societal support built on the back of emotional stories of patient hardship. However, the environment is changing as the collective budget impact of treatments for rare diseases is rising. Despite the provision of alternative funding mechanisms for orphan drugs, payers are taking a tougher line on prices, negotiating rebates, and setting budget impact thresholds. Rather than accepting the labelled indication *de facto*, payers are increasingly scrutinising the patient population as a means of reducing economic uncertainties. In the past, payers always recognised rarity in their pricing decisions. Is this still true or, with the rise of treatments for rare diseases, are other factors contributing more to the price-setting process?

Methodology

Our analysis included 16 orphan drugs approved by the European Medicines Agency (EMA) since July 2012 (see Table 1). We calculated the average annual treatment cost (AATC) of each drug across EU5 (France, Germany, Italy, Spain and the UK) and Japan using launch list prices where possible. While we used list prices, we understand that confidential net price discounts may have been negotiated in France, Italy, Spain and the UK, which would affect actual budget impact.

Discussion and conclusions

We found that there is a relationship between AATC and disease prevalence, in which AATC tends to increase as the prevalence decreases (see Figure 1). While this is true for rare diseases as payers' major focus is on budget impact, the correlation between AATC and prevalence is not as apparent for ultra-rare diseases. Though a trend cannot be established due to the limited number of ultra-rare disease products (n=4), the change in price-prevalence correlation may be symptomatic of a ceiling to payers' willingness to pay.

This study provides evidence that disease rarity may be a driver of price. However, there may be a prevalence threshold below which payer willingness to pay for a given treatment stops increasing. This has important implications for pharmaceutical companies wishing to launch in ultra-rare diseases, potentially including high-profile gene and cell therapies.

Given the stronger relationship between AATC and prevalence seen in existing treatments for less rare diseases, payers will likely continue to expect similar price-prevalence relationships for new entrants. To differentiate on price, manufacturers can look to capitalise on several other factors, including the robustness of clinical data, magnitude of clinical benefit, mortality benefit, level of unmet need, burden of disease and lack of available treatments, and ensure these are captured in their payer negotiation strategies.

In conclusion, payers do value rarity, but need to balance this with the clinical benefit and budget impact uncertainties that products for rare diseases present to ensure optimal price-to-value alignment. Ensuring that patient populations are clearly defined and that the patient journey is well understood will help to reduce some of that uncertainty. Manufacturers should also select indications with the highest clinical benefit and unmet need, and support submissions with compelling patient and/or physician advocacy.

Table 1: Prevalence estimates and sources used

Brand name	Molecule name	EMA approval date	Prevalence	Prevalence sources
Kalydeco	ivacaftor	July, 2012	1:30,000	Maiuri et al., 2015
Opsumit	macitentan	December, 2013	1:5,500	EMA
Adempas	riociguat	March, 2014	1:5,000	EMA
Cerdelga	eliglustat tartrate	January, 2015	1:35,000	EMA
Ofev	nintedanib	January, 2015	1:3,500	EMA
Strensiq	asfotase alfa	August, 2015	1:300,000	Orphanet; Mornet et al., 2011; Conti et al., 2017
Kanuma	sebelipase alfa	August, 2015	1:90,000	Desai et al., 2016, Carter et al., 2019
Raxone	ibedenone	September, 2015	1:35,000	NORD, LHON Society, Man et al., 2002
Orkambi	lumacaftor/ivacaftor	October, 2015	1:5,000	Maiuri et al., 2015[2]
Wakix	pitolisant	April, 2016	1:2,500	Scheer et al., 2018
Uptravi	selexipag	May, 2016	1:5,500	EMA
Galafold	migalastat hydrochloride	May, 2016	1:10,000	EMA
Ocaliva	obeticholic acid	December, 2016	1:2,500	EMA
Spinraza	nusinersen	May, 2017	1:65,000	Verhaart et al., 2017
Brineura	cerliponase alfa	May, 2017	1:2,000,000	Williams et al., 2017
Crysvita	burosumab	February, 2018	1:20,000	Orphanet

Rare Ultra-rare

These drugs have been sorted into two categories: rare and ultra-rare. As proposed by the European Commission, drugs indicated for diseases with a prevalence of less than 1 in 50,000 were categorised as ultra-rare; the remainder, all of which met the EU orphan definition of a prevalence below 5 in 10,000, were categorised as rare. We used a pragmatic approach to calculate prevalence: EMA-prevalence rates served as a foundation for our calculations and were compared against estimates in the literature. Where EMA prevalence differed substantially from the literature, an average was taken from the estimates.

Figure 1: Relationship between average annual treatment cost (AATC) and disease prevalence for a) all diseases and b) diseases with a prevalence of at least 1 in 50,000

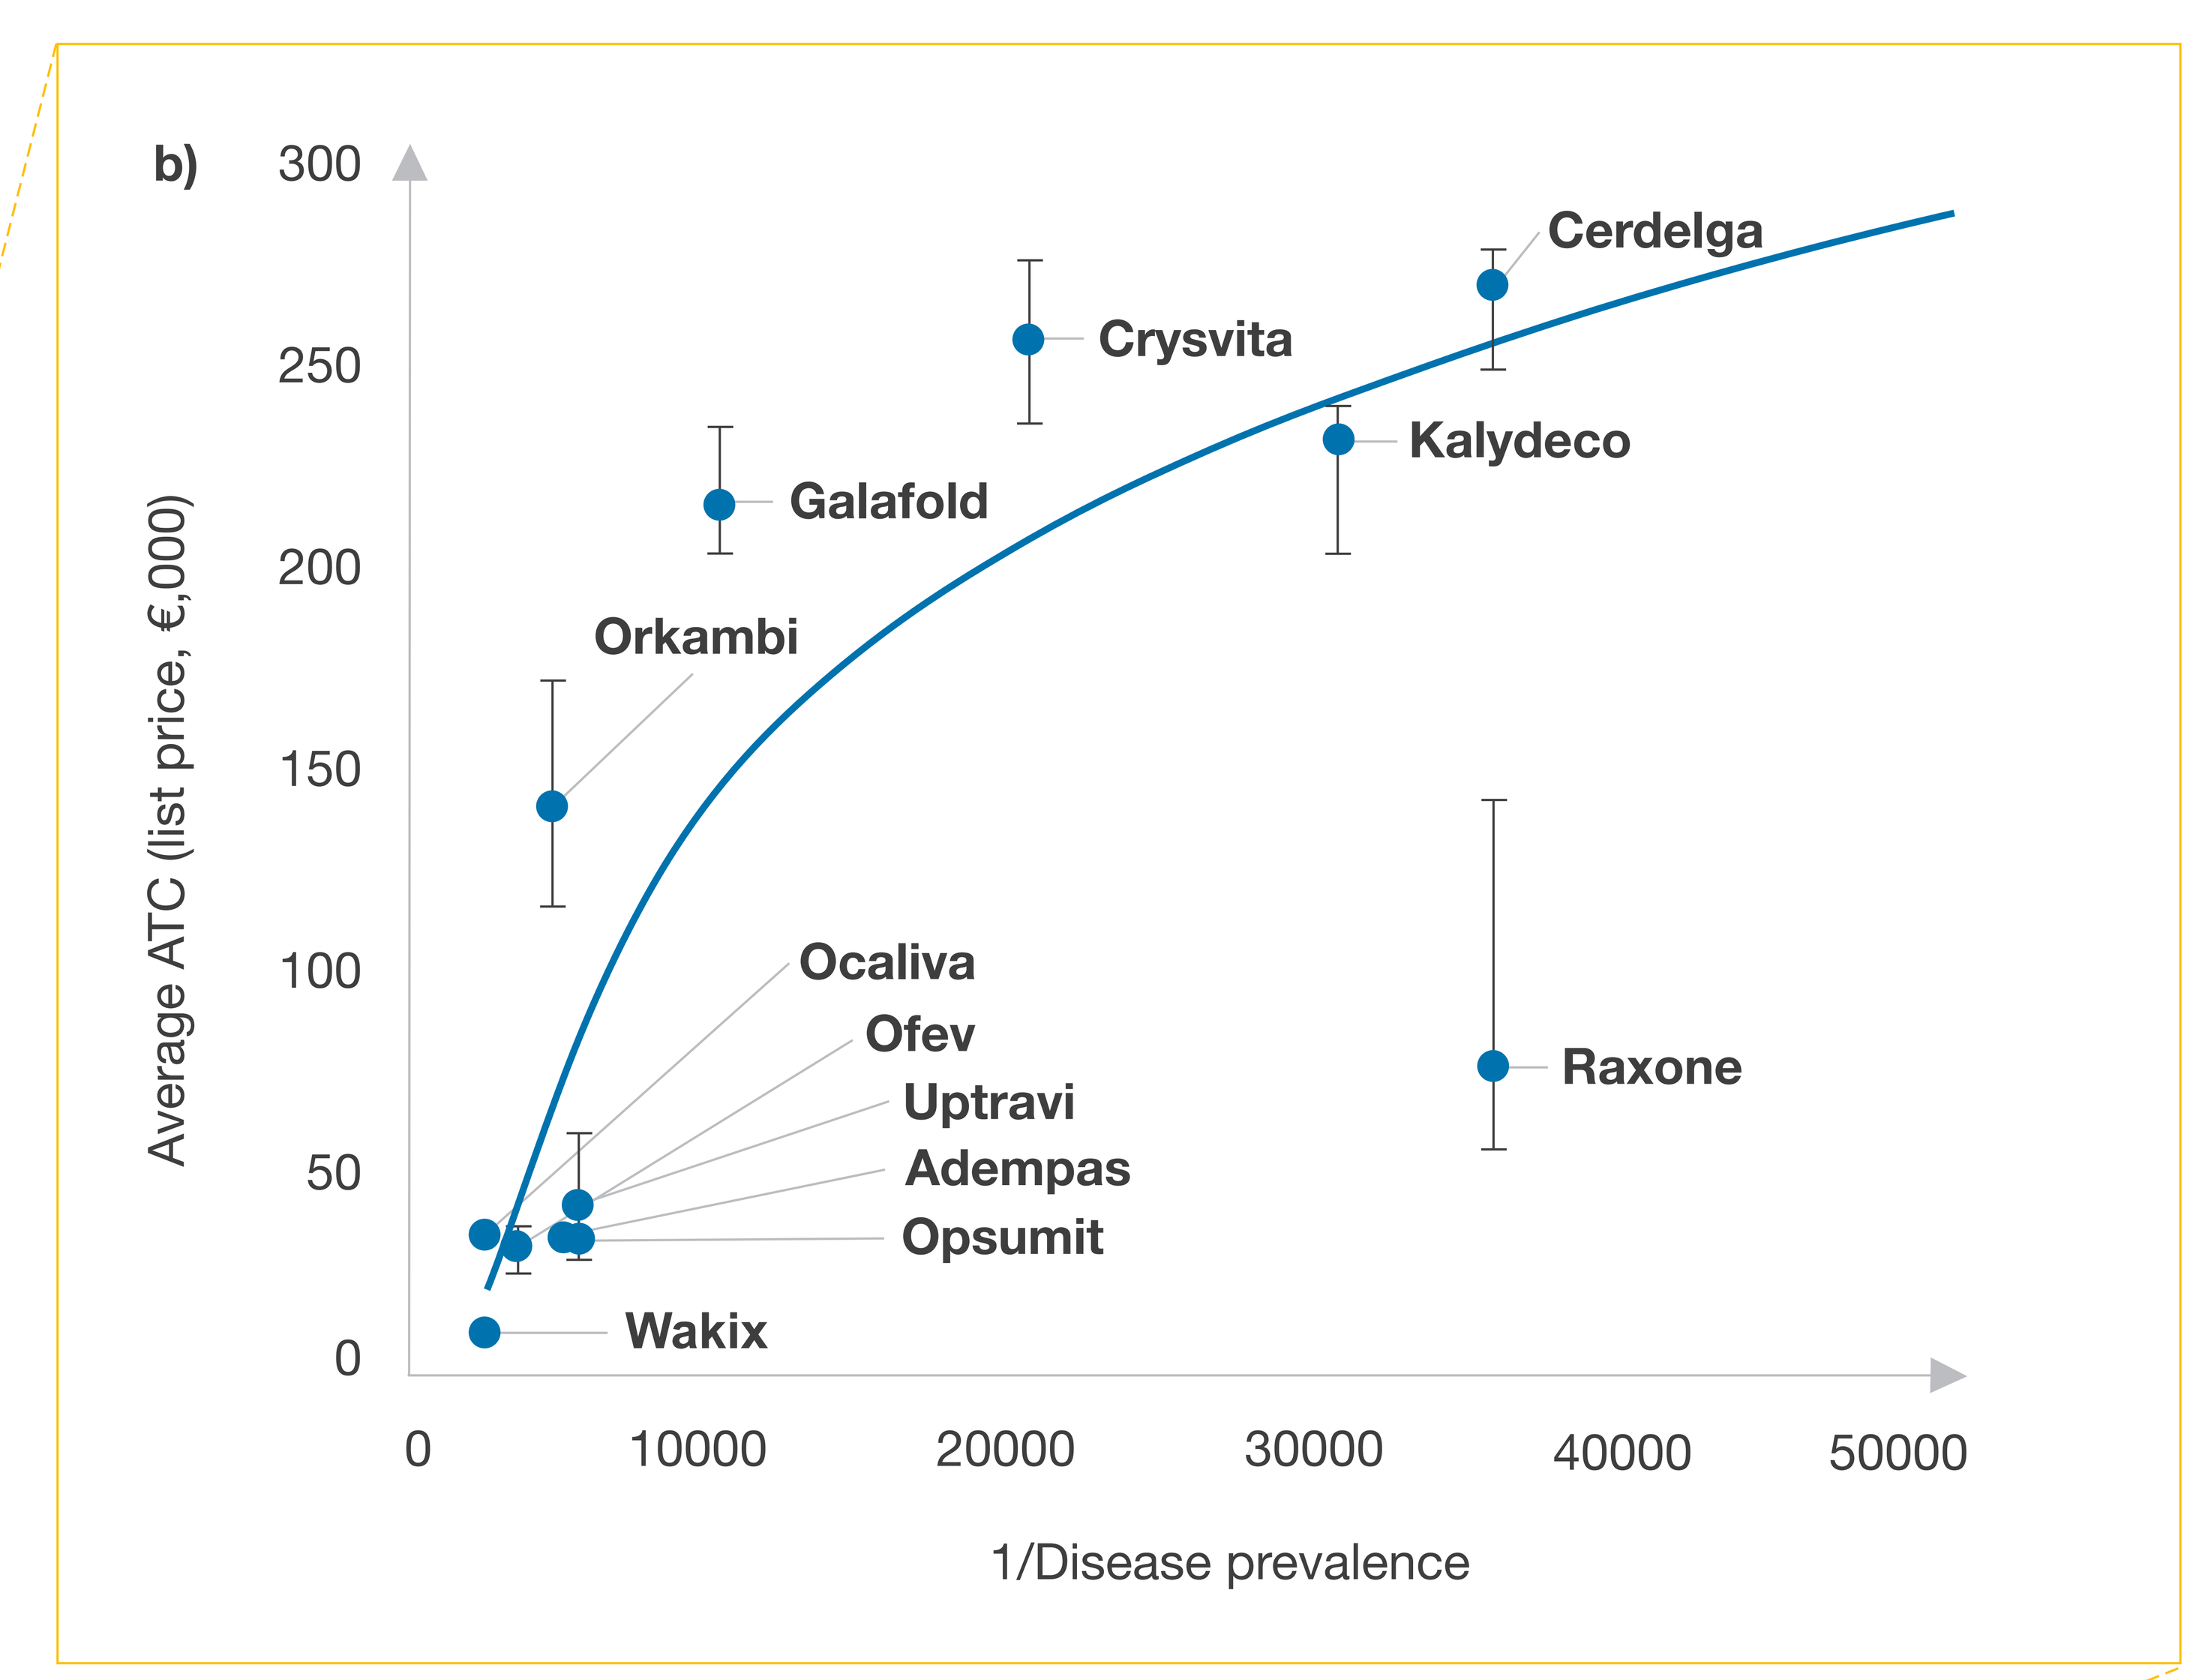
b)
300
250
200
150
100
50
0
Average ATC (list price, €,000)
Cerdelga
Crysvita
Kalydeco
Galafold
Orkambi
Ocaliva
Ofev
Uptravi
Adempas
Opsumit
Raxone
Wakix
0
10000
20000
30000
40000
50000
1/Disease prevalence

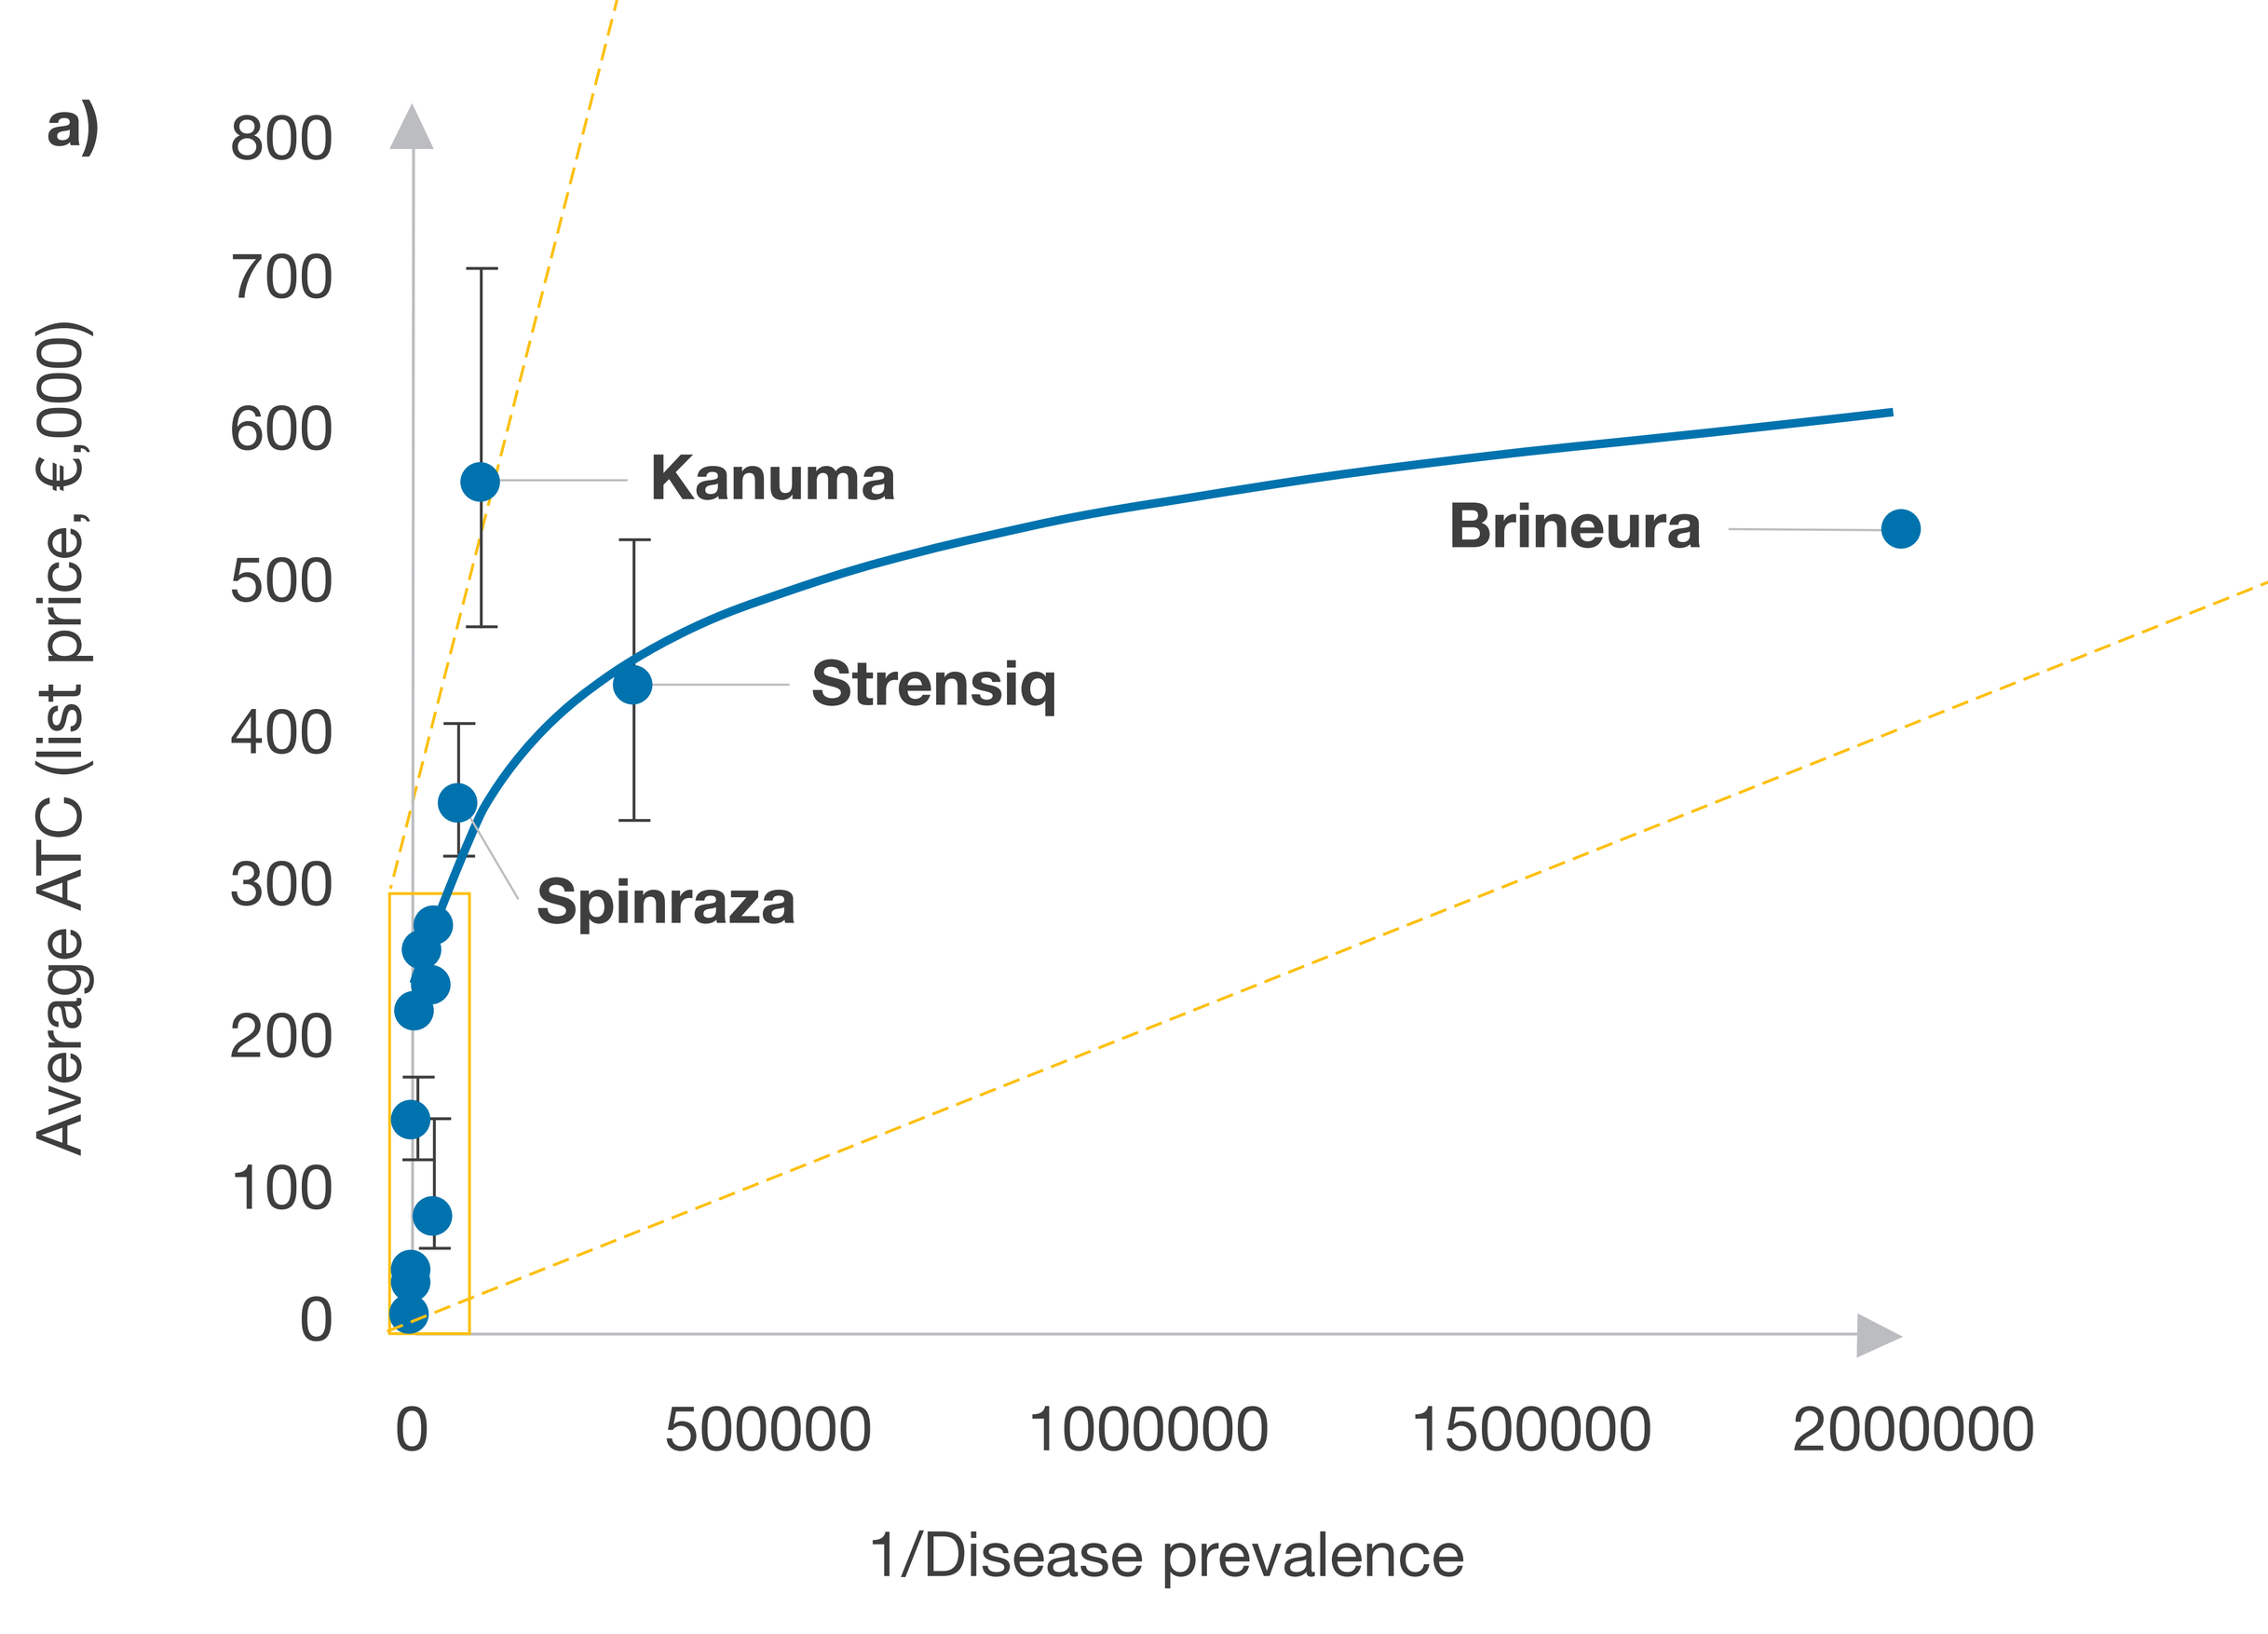
a)
800
700
600
500
400
300
200
100
0
Average ATC (list price, €,000)
Kanuma
Brineura
Strensiq
Spinraza
0
500000
1000000
1500000
2000000
1/Disease prevalence

Our research highlighted that the overall relationship between price and prevalence can be influenced by several other factors including robustness of clinical data, magnitude of clinical benefit, demonstrated mortality benefit, level of unmet need, disease burden and lack of available treatments.

We also found that epidemiology estimates could vary across markets when local data sources were used in HTA evaluations. This variability results in different quantifications of the eligible patient population, which could be leading to price disparities across markets.

The views expressed here belong to the authors and do not represent the views of Charles River Associates (CRA).

Charles River Associates